PROcedure-SPECific postoperative pain managemenT (PROSPECT) recommendations: a rigorous methodology to minimise bias

prospect

procedure specific postoperative pain management

Edmund Neugebauer[1] and Stephan A Schug[2], on behalf of the PROSPECT Working Group

[1]Professor, Biochemical and Experimental Division, Medical Faculty, University of Cologne, Cologne, Germany; [2]Professor, UWA Anaesthesia, School of Medicine and Pharmacology, University of Western Australia, Perth, Western Australia

Background and Aims

- PROSPECT: web-based clinical decision support programme, which aims to formulate robust evidence-based recommendations for procedure-specific postoperative pain management.
- Initiated by an expert Working Group of surgeons and anaesthesiologists.
- The PROSPECT methodology has been refined, with the aim of increasing the rigour and transparency of the systematic review process and of the formulation of the consensus recommendations by the Working Group.

Review of abstracts and identified papers

Inclusion criteria:

- Randomised clinical trials of analgesic, anaesthetic and operative interventions relating to the surgical procedure being reviewed
- Pain scores from a linear pain scale, e.g. visual analogue scale (VAS) or verbal or numerical rating scale (VRS, NRS)
- Only English language studies are included, due to time and resource constraints

Subgroup (consists of at least two members of the Working Group): agrees literature search terms, determines the level of evidence of each study, reviews the summary 'Outcomes Document', drafts transferable evidence, clinical practice information and recommendations

Methods

- The refined methodology was formulated by the Working Group and first applied in the 2006 updated review of postoperative pain management for laparoscopic cholecystectomy.

Assessment of quality and level of evidence of each study

The researcher performs quality assessment:

– quality scores are assigned, according to Jadad *et al.*,[1] to indicate appropriate randomisation (0–2), blinding (0–2) and statement of withdrawals (0–1)

– scores A–D are assigned to indicate adequacy of allocation concealment: A=adequate, B=unclear, C=inadequate; D=not used

The quality assessment is validated by the PROSPECT Subgroup. The level of evidence is then determined by the PROSPECT Subgroup and the researcher independently, according to Sackett *et al.*[2] Oxford Centre for Evidence-Based Medicine tables.

Results

- The step-by-step process by which the PROSPECT Working Group formulates its recommendations is presented in the Procedural Flowchart.

Procedural Flowchart

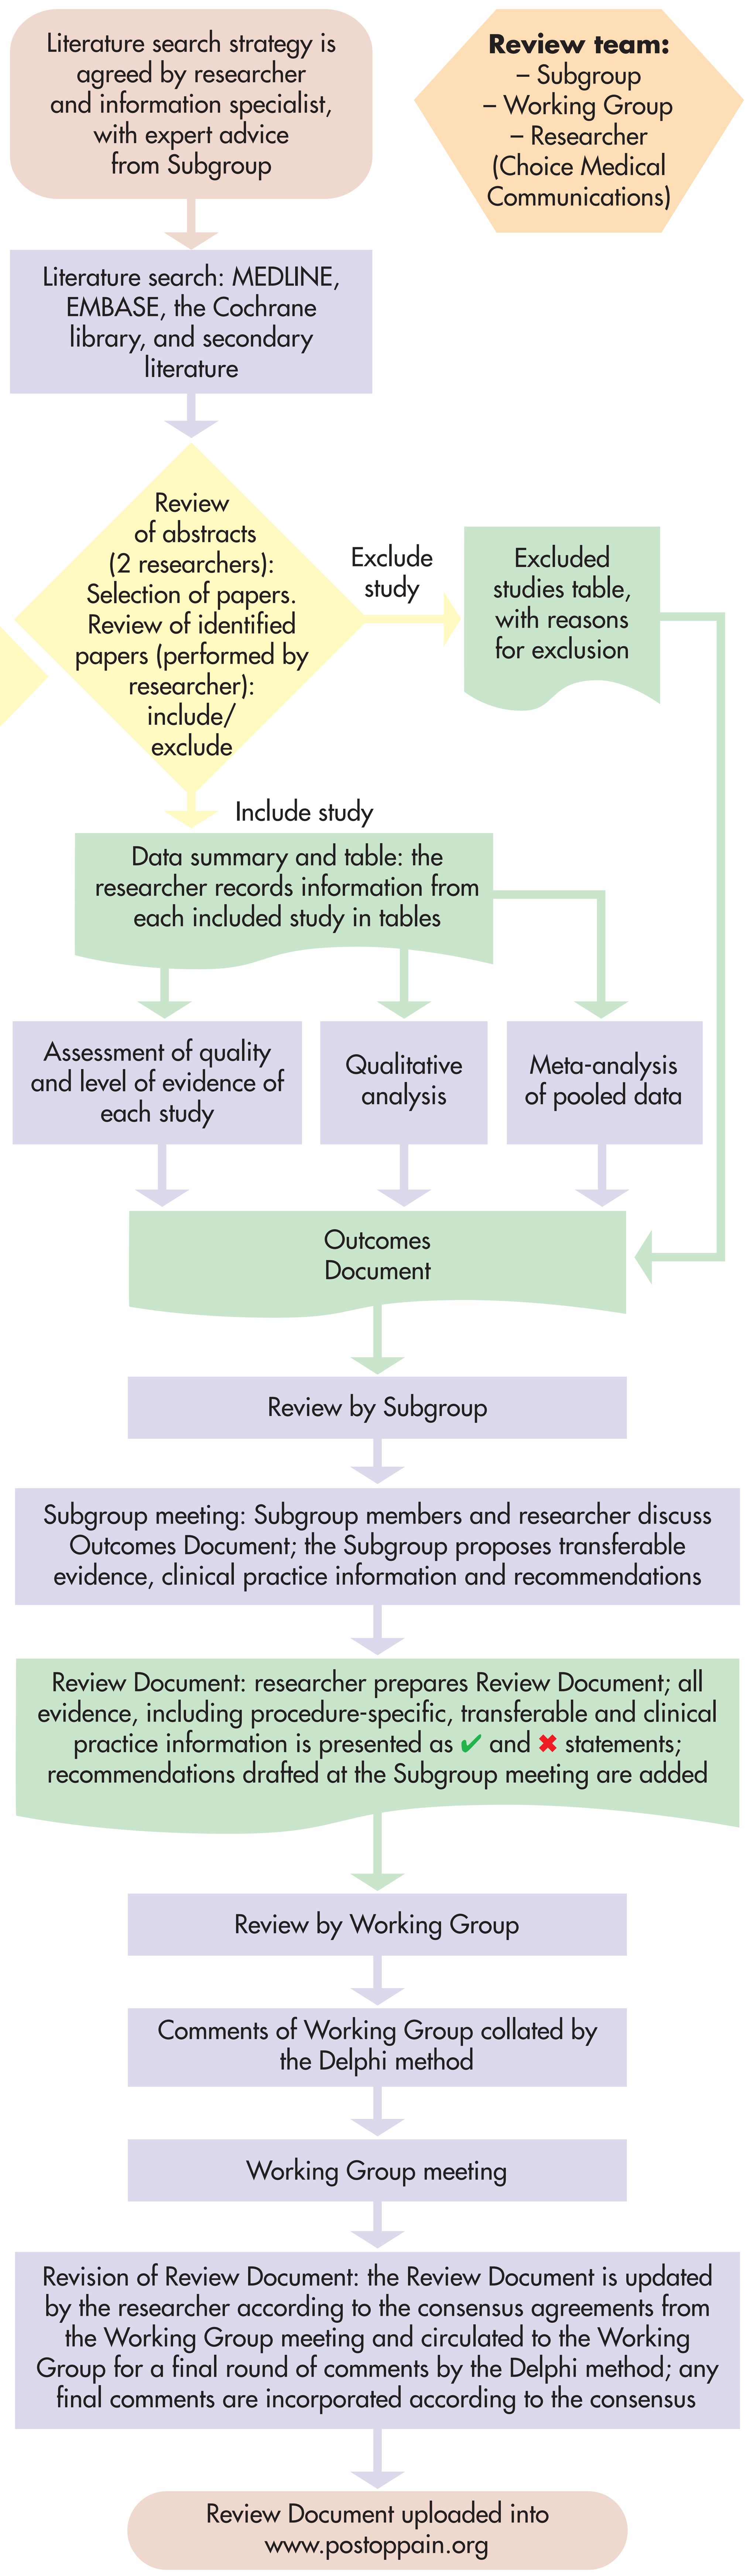
Literature search strategy is agreed by researcher and information specialist, with expert advice from Subgroup
Review team: – Subgroup – Working Group – Researcher (Choice Medical Communications)
Literature search: MEDLINE, EMBASE, the Cochrane library, and secondary literature
Review of abstracts (2 researchers): Selection of papers. Review of identified papers (performed by researcher): include/exclude
Exclude study
Excluded studies table, with reasons for exclusion
Include study
Data summary and table: the researcher records information from each included study in tables
Assessment of quality and level of evidence of each study
Qualitative analysis
Meta-analysis of pooled data
Outcomes Document
Review by Subgroup
Subgroup meeting: Subgroup members and researcher discuss Outcomes Document; the Subgroup proposes transferable evidence, clinical practice information and recommendations
Review Document: researcher prepares Review Document; all evidence, including procedure-specific, transferable and clinical practice information is presented as ✔ and ✖ statements; recommendations drafted at the Subgroup meeting are added
Review by Working Group
Comments of Working Group collated by the Delphi method
Working Group meeting
Revision of Review Document: the Review Document is updated by the researcher according to the consensus agreements from the Working Group meeting and circulated to the Working Group for a final round of comments by the Delphi method; any final comments are incorporated according to the consensus
Review Document uploaded into www.postoppain.org

Qualitative analysis

The researcher performs qualitative analyses for each group of studies that report similar treatment comparisons (for example, all studies comparing incisional local anaesthetics *versus* placebo); all analyses are reported in the summary Outcomes Document

Meta-analysis of pooled data

The researcher performs quantitative analyses where possible, using Review Manager 4.2.2 software,[3] which has been developed for Cochrane Collaboration systematic reviews; quantitative analyses are reported in the summary Outcomes Document

Working Group process: formulation of consensus recommendations

Delphi method

Following evaluation of the Review Document by the Working Group members, comments on the evidence and recommendations are forwarded only to the researcher, and not to the whole Working Group; individual comments are then collated for discussion at a round-table meeting

Working Group meeting

Collated comments are discussed and the recommendations are finalised; where consensus is not reached by group discussion, a modified Nominal Group Process is used, which involves iterative rounds of discussion and voting, until consensus is reached

Conclusion

- The process by which the PROSPECT Working Group formulates its recommendations has been refined to take greater account of the quality of available evidence and to reduce the potential for bias in the formulation of recommendations. This improved methodology should help fulfil the core aims of PROSPECT and give users added confidence in the evidence-based recommendations.

References

1. Jadad AR, *et al. Control Clin Trials* 1996; **17**: 1–12.
2. Sackett DL, *et al. Evidence-Based Medicine: How to Practice and Teach EBM*. London: Churchill Livingstone, 2000.
3. RevMan Analyses [Computer program]. Version 1.0 for Windows. In: *Review Manager (RevMan) 4.2. Oxford, England: The Cochrane Collaboration*, 2003

Acknowledgement

PROSPECT is supported by Pfizer.